A Phase I and Pharmacokinetic (PK) Trial of AP5346, a Novel Polymer-Linked DACH Platinum (Pt) Compound, Administered as an Intravenous (IV) Infusion Once a Week for 3 Consecutive Weeks in Advanced Solid Tumor Patients

Mario Campone [1], Jeany M Rademaker-Lakhai [2], Jaafar Bennouna [1], Stephen B Howell [3], Richard F de Boer [4], David P Nowotnik [5], Jos H Beijnen [2], Jan HM Schellens [2]

[1] Centre René Gauducheau, Nantes, France; [2] The Netherlands Cancer Institute, Amsterdam, The Netherlands; [3] University of California Cancer Center, San Diego, USA;
[4] NDDO Oncology B.V., Amsterdam, The Netherlands; [5] Access Pharmaceuticals, Inc., Dallas, USA

1. Background

AP5346 is a macromolecular carrier linked to a bioactive DACH-platinum complex. The macromolecular carrier is a water soluble biocompatible 90:10 random copolymer of a N-(2-hydroxypropyl)methacrylamide (HPMA) monomer and a methacrylamide monomer substituted with a tri-glycine group, the linker to the platinum complex (Figure 1).

AP5346 was designed to take advantage of the enhanced permeability and retention effect, in which tumor neovasculature shows greater permeability to macromolecules than normal tissue, often associated with limited lymphatic and/or capillary drainage.

The DACH-platinum complex is thought to be biologically inert while attached to the polymer, with slow in vivo release accelerated by low pH environments, such as the extracellular space of hypoxic tumors and the intracellular lysomal compartment.

AP5346 produced greater tumor growth inhibition and prolonged growth delay compared to equitoxic oxaliplatin and/or carboplatin doses in B16 melanoma, 2008 ovarian, HT-29 colorectal, and Lewis lung tumor models.

We have undertaken the first phase I trial in solid tumor patients of AP5346 administered weekly over 1 hour, for 3 weeks out of every 4 weeks.

2. Study Objectives

Primary Objectives

- To determine the maximum tolerated dose (MTD) of a course of 3 weekly 1 hr infusions of AP 5346 given on days 1, 8 and 15 every 4 weeks;
- To establish a recommended dose for phase II trials on this dose schedule.
- To determine the qualitative and quantitative toxic effects of 3 weekly 1 hr infusions of AP 5346 and to study the predictability, duration, intensity, onset, reversibility and dose-relationship of the toxic side effects;

Secondary Objectives

- To determine the pharmacokinetics of total and ultrafiltrable platinum and circulating WBCs DNA adduct levels as a function of AP 5346 dose.
- To document possible antitumor activity.
- To measure the level of platinum-DNA adducts in tumor sample 24 hours after the first dose of AP 5346.

Figure 1. Partial chemical structure of AP5346, a random copolymer, where q:r:s is approximately 36-47 : 2.3 : 1

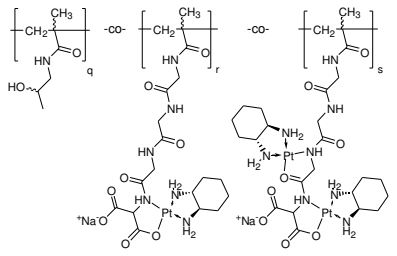

3. Study Design

Open label, dose-finding phase I study conducted in 2 centers in France and Holland

Administration schedule

AP5346 was administered in a 1-hour IV infusion in 500 mL of 5% glucose once per week for 3 weeks with a 1 week rest.

Dosing on days 8 or 15 was omitted in case of grade 3 thrombocytopenia, neutropenia, or leukopenia

Anti-emetic prophylaxis was not mandated by the protocol, but, from DL 5, 5-HT3 antagonists and corticosteroids were administered prophylactically to most patients.
The following hydration and urine alkalization scheme was applied in DL 5-7

Prehydration (immediately prior to infusion)	Na bicarbonate 2000 mL 1.4% IV 12 hr
Posthydration (immediately post-infusion)	Na bicarbonate 2000 ml L.4% IV 20-22 hr
Day 2 until next infusion	Na bicarbonate 1000 mg p.o. 4 x daily
Rescue if pH < 7 in 24 hours after infusion	Na bicarbonate 100 mL 4,2% or 50 mL 8.4% IV

Planned Dose Escalation Scheme

The starting dose was 40 mg Pt/m², one-tenth of the MTD in the most sensitive species (female mice). The magnitude of dose escalation with successive dose levels was determined according to the severity of toxicity in preceding DL, as follows:

Maximum NCI-CTC grade of toxicity in prior DL: Hematologic[a]		Non-hematologic[b]	Dose Escalation	Initial number of patients per DL
0-1	and	0	100%	1
2	or	1	50%	3
3-4	or	2-4	20% - 33%	3

[a] except anemia [b] except alopecia and untreated nausea/vomiting

In the event of DLT during cycle 1, a total of 6 patients were to be entered in that dose level.

Definition of DLT

DLT was defined as any of the following treatment-related toxicities:

- Non-hematologic toxicity grade 3-4 (except inadequately treated nausea/vomiting);
- Neutrotoxicity grade 2 lasting > 2 weeks.
- Neutropenia grade 4 lasting > 5 days or complicated by fever or infection;
- Thrombocytopenia grade 4;
- The inability to receive 3 weekly consecutive doses of the drug for reasons of toxicity which in itself are not dose-limiting, but might become dose-limiting if administrations would continue
- Delay of cycle 2 by > 2 weeks

Definition of MTD

The MTD was defined as the DL in which $\geq$ 2/6 patients experienced a DLT during the first cycle of therapy.

The RD was to be one dose level below the MTD

4. Principal Eligibilty Criteria

- Histological or cytologically confirmed advanced solid tumor not amenable to standard treatment
- WHO Performance Status 0-2
- Age $\geq$18 years
- Adequate hematological and biochemical function
 - Bone marrow: Absolute neutrophil count (ANC) $\geq$ 1.5 x 109/L; platelet count $\geq$ 100 x 109/L
 - Hepatic: Total serum bilirubin $\geq$ 1.5 x the upper limit of normal (ULN); alkaline phosphatase (AP), AST and ALT $\geq$ 3 x ULN
 - Renal: serum creatinine, blood urea $\geq$ 1 x ULN, calculated creatinine clearance > 50 mL/min (Cockroft and Gault)
- Absence of clinical signs of brain metastases and/or leptomeningeal involvement
- Neuropathy $\geq$CTC grade 2, or hearing loss $\geq$ CTC grade 2.
- No prior nephrectomy.
- Signed informed consent

5. Pharmacokinetics

Pharmacokinetic samples were taken in all treated patients on day 1 of cycle 1, at the following time points: prior to infusion, and 0, 15, 30 min, 1, 2, 4, 7, 24, 48, 96 hours after the end of the infusion. Samples were also taken prior to each infusion during first 2 cycles to measure platinum trough levels.

Plasma samples were analyzed by atomic absorption spectrophotometry for total platinum and ultrafiltrable platinum (<3 kDa), with a lower limit of quantitation of 0.1 µg/mL and 0.02 µg/mL, respectively.

Pharmacokinetic parameters were calculated using WinNonlin ver 3.1 by non-compartmental methods .

6. Results

Between April 2003 and December 2004, 26 patients were treated in 7 dose levels

Dose level	Dose per infusion (mg Pt/m²)	Number of patients: Treated	Evaluable for MTD	Evaluable for safety	Evaluable for efficacy
1	40	1	1	1	1
2	80	1	1	1	-
3	160	3	2	3	2
4	320	3	3	3	2
5	640	6	6	6	4
6	850	6	5	6	4
7	1280	6	6	6	4
Total		26	24	26	17

Patient Characteristics

	DL 1-3	DL 4	DL 5	DL 6	DL 7	Total
Number of patients treated	5	3	6	6	6	26
Sex						
Female	4	1	3	3	4	14
Male	2	2	3	3	2	12
Median age (range)	60 (33-72)	47 (39-51)	58 (43-70)	48 (32-50)	64 (32-74)	50 (32-74)
Performance status (WHO)						
0	-	2	5	1	3	11
1	4	1	-	5	3	13
2	1	-	1	-	-	2
Histology						
Melanoma		1	1	1	2	5
Breast	3		1			4
Ovarian		1	1		2	4
Other[a]	2	1	3	5	2	13

	DL 1-3	DL 4	DL 5	DL 6	DL 7	Total
N prior chemotherapy regimens	2 (1-4)	1 (1-3)	1 (0-3)	2 (0-3)	1.5 (1-3)	1.5 (0-3)
No. involved sites	4 (1-4)	4 (2-4)	3 (1-5)	3.5 (2-5)	3 (2-4)	3 (1-5)
Involved sites						
Lymph Nodes	3	3	4	4	4	18
Lung	2	2	3	5	5	17
Liver	1	1	4	2	3	11
Bone	2		4	1	1	8
Primary Tumor	1	1	2	3	1	8
Other[b]	5	3	2	6	4	20

[a] ACUP, cervix, esophageal, NSCLC, pancreatic, prostate, renal, thyroid, Ewing sarcoma, urachus, peritoneal carcinoma
[b] Pleural effusion, skin, CNS, pericardial effusion, adrenal, kidney, mediastinum, mesenterium.

Treatment exposure and reasons for discontinuation

	DL 1-3 N	DL 4 N	DL 5 N	DL 6 N	DL 7 N	Total N	%
Number of patients treated	5	3	6	6	6	26	
Cycles administered	8	5	13	6	9	41	
Median n cycles per patient	2 (1-2)	2 (1-2)	2 (1-4)	1 (1)	1 (1-3)	1 (1-4)	
Reason for discontinuation							
Progressive disease	3	1	3	3	1	11	42
AE: Treatment related		1	3	1	4	9	35
AE: treatment unrelated	2			1		3	12
Patient refusal				1	1	2	8
Investigator decision		1				1	4

Treatment toxicity resulted in 4 cycle delays (DL 1, 5, 7), the omission of 6 infusions (DL 3, 5, 7) and 2 dose reductions (DL 7)

7. Dose Escalation and Determination of MTD

Dose escalation proceeded by 100% dose increments through DL 1 to 5 (640 mg Pt/m²) and 7 (1280 mg Pt/m²). An intermediate DL 6 (850 mg Pt/m²) was subsequently explored.

No DLT were observed during the first cycle in dose levels 1-4

DL 6 was MTD with DLT during the first cycle in 2 out of 5 patients

DL 7 was toxic with DLT in 5 out 6 patients having a DLT in first cycle

DL 5 is the recommended dose, 1 patient out of 6 having experienced anuria grade 3 accompanied by creatinine elevation grade 4

Dose-limiting toxicity

Dose level	N patients evaluable	N patients with DLT	Main toxicities
5 (RD)	6	1	Creatinine elevation gr 4 Anuria gr 3
6 (MTD)	5	2	Fatigue gr 3
			Vomiting gr 3
7	6	5	Neutropenic infection gr 4 Diarrhea gr 3
			Fatigue gr 3
			Hypophosphatemia gr 4 Bicarbonate gr 3 Renal insufficiency gr 2 Fatigue gr 3 Nausea/ vomiting gr 3 Neutropenia gr 4 Spastic ileus gr 3
			Fatigue gr 3 Nausea/ vomiting gr 3

8. Safety

Two deaths occurred at DL 5 due to renal failure, possibly drug-related. One had not received hydration / urine alkalization prophylaxis and the other had right renal involvement with left renal vein thrombosis

Seven (7) patients (27%) experienced treatment-emergent grade 1-2 creatinine elevation or clearance decrease, without clinical signs of renal dysfunction. IV urine alkalization appeared to improve tolerance, with 0% vs 46% grade 2-4 renal toxicity in patients with/ without prophylaxis in DL 5-7

70% of patients experienced vomiting, reaching grade 3-4 in 5 patients in DL 5-7 (28%), of whom only 1 had received adequate anti-emetic prophylaxis.

Three (3) patients (12%) reported treatment-emergent grade 1 sensory neuropathy of 1 day duration; no ototoxicity occurred

One (1) case of anaphylactic shock (during the 5th infusion) and a case of urticaria grade 2 (during the 3rd infusion) led to study treatment discontinuation

Until DL 7, neutropenia did not occur and thrombocytopenia was sporadic

Toxicity per patient according to dose level

	DL 1-4 (N=8) Gr 1-4	DL 1-4 Gr 3-4	DL 5 (N=6) Gr 1-4	DL 5 Gr 3-4	DL 6 (N=6) Gr 1-4	DL 6 Gr 3-4	DL 7 (N=6) Gr 1-4	DL 7 Gr 3-4
Neutropenia	-	-	-	-	-	-	2	2
Thrombocytopenia	1	-	3	-	1	-	4	-
Creatinine increased	-	-	3	1	1	-	4	-
Creatinine clearance decreased	1	-	2	2	-	-	5	-
Fatigue	1	-	3	1	1	1	3	1
Nausea	4	-	5	1	4	-	6	2
Vomiting	8	-	1	1	4	1	5	3
Abdominal pain	2	-	1	-	-	-	1	-
Anorexia	2	-	2	1	1	-	1	-
Diarrhoea	2	1	2	-	-	-	2	1
Ileus (spastic)	-	-	1	1	1	-	1	1
Stomatitis	1	-	1	-	1	-	1	-

Figure 2. Evolution of calculated creatinine clearance during treatment in DL 5-7 in each patient (% change with respect to baseline)

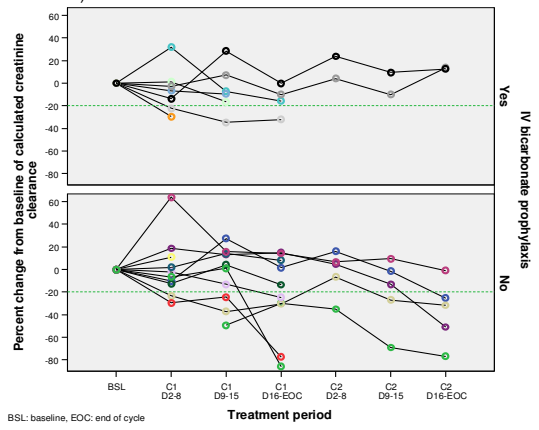
Percent change from baseline of calculated creatinine clearance
IV bicarbonate prophylaxis
Yes
No
BSL
C1 D2-8
C1 D9-15
C1 D16-EOC
C2 D2-8
C2 D9-15
C2 D16-EOC
Treatment period

BSL: baseline, EOC: end of cycle

9. PK Results

- PK results are available for the first 18 patients
- Cmax and AUC0-1wk increased linearly with dose for total plasma platinum (r^2 = 0.92 and 0.85), and were probably linear for ultrafiltrate platinum (r^2 = 0.89 and 0.64)
- Ultrafiltrate platinum AUC was <1% of total platinum AUC, suggesting that only a small fraction of platinum is released in plasma as small molecular entities
- Terminal half-life did not appear to vary with dose. Mean terminal half-life was 68.6 $\pm$ 19.3 hours and 57.7 $\pm$ 27.1 hours for total and ultrafiltrate platinum, respectively. Ultrafiltrate half-life is longer than those reported for cisplatin (0.7-0.8 hr), carboplatin (1.7-5.9 hr) and oxaliplatin (24-27 hr) [1].
- There was no obvious effect on PK parameters in the 5 patients receiving prophylactic hydration and urine alkalization

Dose (mg Pt/m²)	N patients	Total plasma platinum C_{max} (µg/mL) Mean	SD	$AUC_{0-1\,wk}$ (µg.hr/mL) Mean	SD	Plasma ultrafiltrate platinum C_{max} (ng/mL) Mean	SD	$AUC_{0-1\,wk}$ (ng.hr/mL) Mean	SD
40	1	11.7	-	108	-	185	-	391	-
80	1	14.0	-	119	-	31	-	68	-
160	3	41.0	12.1	352	46	86	33	1571	382
320	3	58.1	13.1	606	144	157	60	3808	2562
640	3	120.0	32.8	1612	763	274	54	15 629	10 938
850	2	156.5	-	1549	-	385	-	8907	-
1280	6	292.0	50.1	3992	787	577	95	26 419	11 320

Figure 3. Mean total platinum plasma concentrations by dose

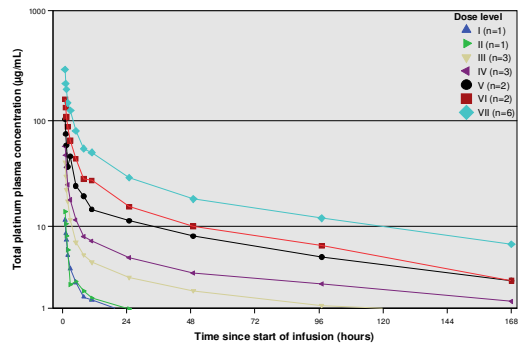
Dose level
I (n=1)
II (n=1)
III (n=3)
IV (n=3)
V (n=2)
VI (n=2)
VII (n=6)
Total platinum plasma concentration (µg/mL)
Time since start of infusion (hours)

Figure 4. Mean ultrafiltrate platinum plasma concentrations by dose level after first infusion

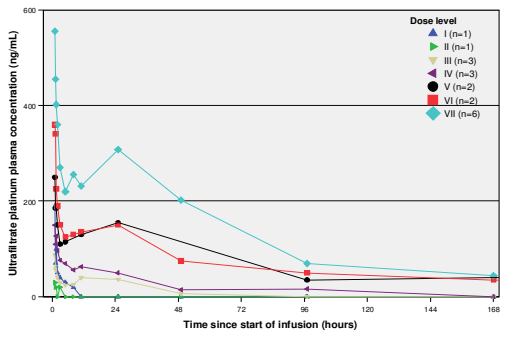
Dose level
I (n=1)
II (n=1)
III (n=3)
IV (n=3)
V (n=2)
VI (n=2)
VII (n=6)
Ultrafiltrate platinum plasma concentration (ng/mL)
Time since start of infusion (hours)

10. Antitumor Activity

Two partial responses (RECIST criteria) were achieved, in a heavily-pretreated, platinum-sensitive ovarian cancer patient and in a dacarbazine-pretreated melanoma patient. An ACUP of suspected ovarian origin who was inevaluable due to discontinuation after 2 infusions, had normalization of CA 125 (from 133 IU/L), with subsequent sustained response to oxaliplatin with a 52% decrease CA 125 from baseline 83 IU/L

Response	DL	Age (years)	Sex	PS	Primary Site	Prior chemotherapy lines	Metastatic sites	N cycles	TTP (months)
PR[a]	5	70	F	0	Ovarian	4	liver, LN, ovary	1	3.3
PR	7	74	M	0	Melanoma	1	lung, LN, skin	1	3.0
SD	4	47	M	1	Esophageal	6	lung, LN, peritoneum, esophagus	2	3.3
SD	5	43	M	0	Melanoma	1	bone, CNS, liver, lung, LN	4	2.8
SD	7	32	F	0	Cervix	1	lung, LN	1	1.8+

11. Conclusions

- The recommended dose for further assessment of AP5346 administered over 1 hour for 3 weeks out of every 4 weeks is 640 mg Pt/m²
- AP5346 was tolerated up to a dose of 640 mg Pt/m², with a toxicity profile characterized by frequent grade 1-2 nausea, vomiting, and creatinine elevations, with an absence of grade 3-4 hematotoxicity below 1280 mg Pt/m².
- Evidence of anti-tumor activity was observed from 640 mg Pt/m², with partial responses achieved in patients with melanoma and ovarian cancer, and disease stabilizations in patients with melanoma, esophageal cancer, and cervix cancer.
- Total and ultrafiltrate platinum Cmax and AUC increased linearly with dose, and terminal half-life did not vary with dose
- Phase II assessment of AP5346 is planned in pretreated platinum sensitive advanced ovarian cancer patients

12. Reference

1. Johnson S, O'Dwyer P. Cisplatin and its analogues. in: DeVita VT, Hellman S, Rosenberg S, editors, Cancer. Principles & practice of oncology. 7th Philadelphia, PA: Lippincott Williams & Wilkins; 2005, p. 344-58.